MBRC-101: a novel antibody-drug conjugate (ADC) targeting the membrane-associated tyrosine kinase receptor EphA5 in breast cancer

Fernanda I. Staquicini[1], Fenny H. F. Tang[2, 3], Vanessa de Oliveira[1], Daniela I. Staquicini[2, 3], Yongjian Wu[1], Kirstin F. Barnhart[1], J. Kellogg Parsons[1], Wadih Arap[2, 4], Isan Chen[1], Renata Pasqualini[2, 3]

[1] MBrace Therapeutics, Inc. San Diego, CA, USA

[2] Rutgers Cancer Institute of New Jersey and Rutgers New Jersey Medical School, Newark, NJ, USA

[3] Division of Cancer Biology, Department of Radiation Oncology; [4] Division of Hematology/Oncology, Department of Medicine

San Antonio Breast Cancer Symposium® - December 5-9, 2023

Introduction

MBRC-101 is a potent EphA5-directed ADC for the treatment of advanced solid tumors

• EphA5 is a member of the receptor tyrosine kinase family of proteins with functional roles in axonal guidance during embryonic development[1,2,3]

• EphA5 expression has been reported in lung, gastric, ovarian and pancreatic cancers, with restricted expression in normal tissues[4,5,6]

• Differential expression in cancers and accessibility through the systemic circulation support the development of antibody-based therapies against EphA5[7,8]

EphA5 is expressed in breast cancer

• Archival breast cancer tissue sections were evaluated by immunohistochemistry (IHC)

• Robust and selective EphA5 expression was detected in > 80% of triple negative breast cancer (TNBC) and > 80% of hormone receptor-positive (HR+) breast cancer tissue samples

• EphA5 expression was not detected in adjacent, non-malignant breast tissue or normal breast

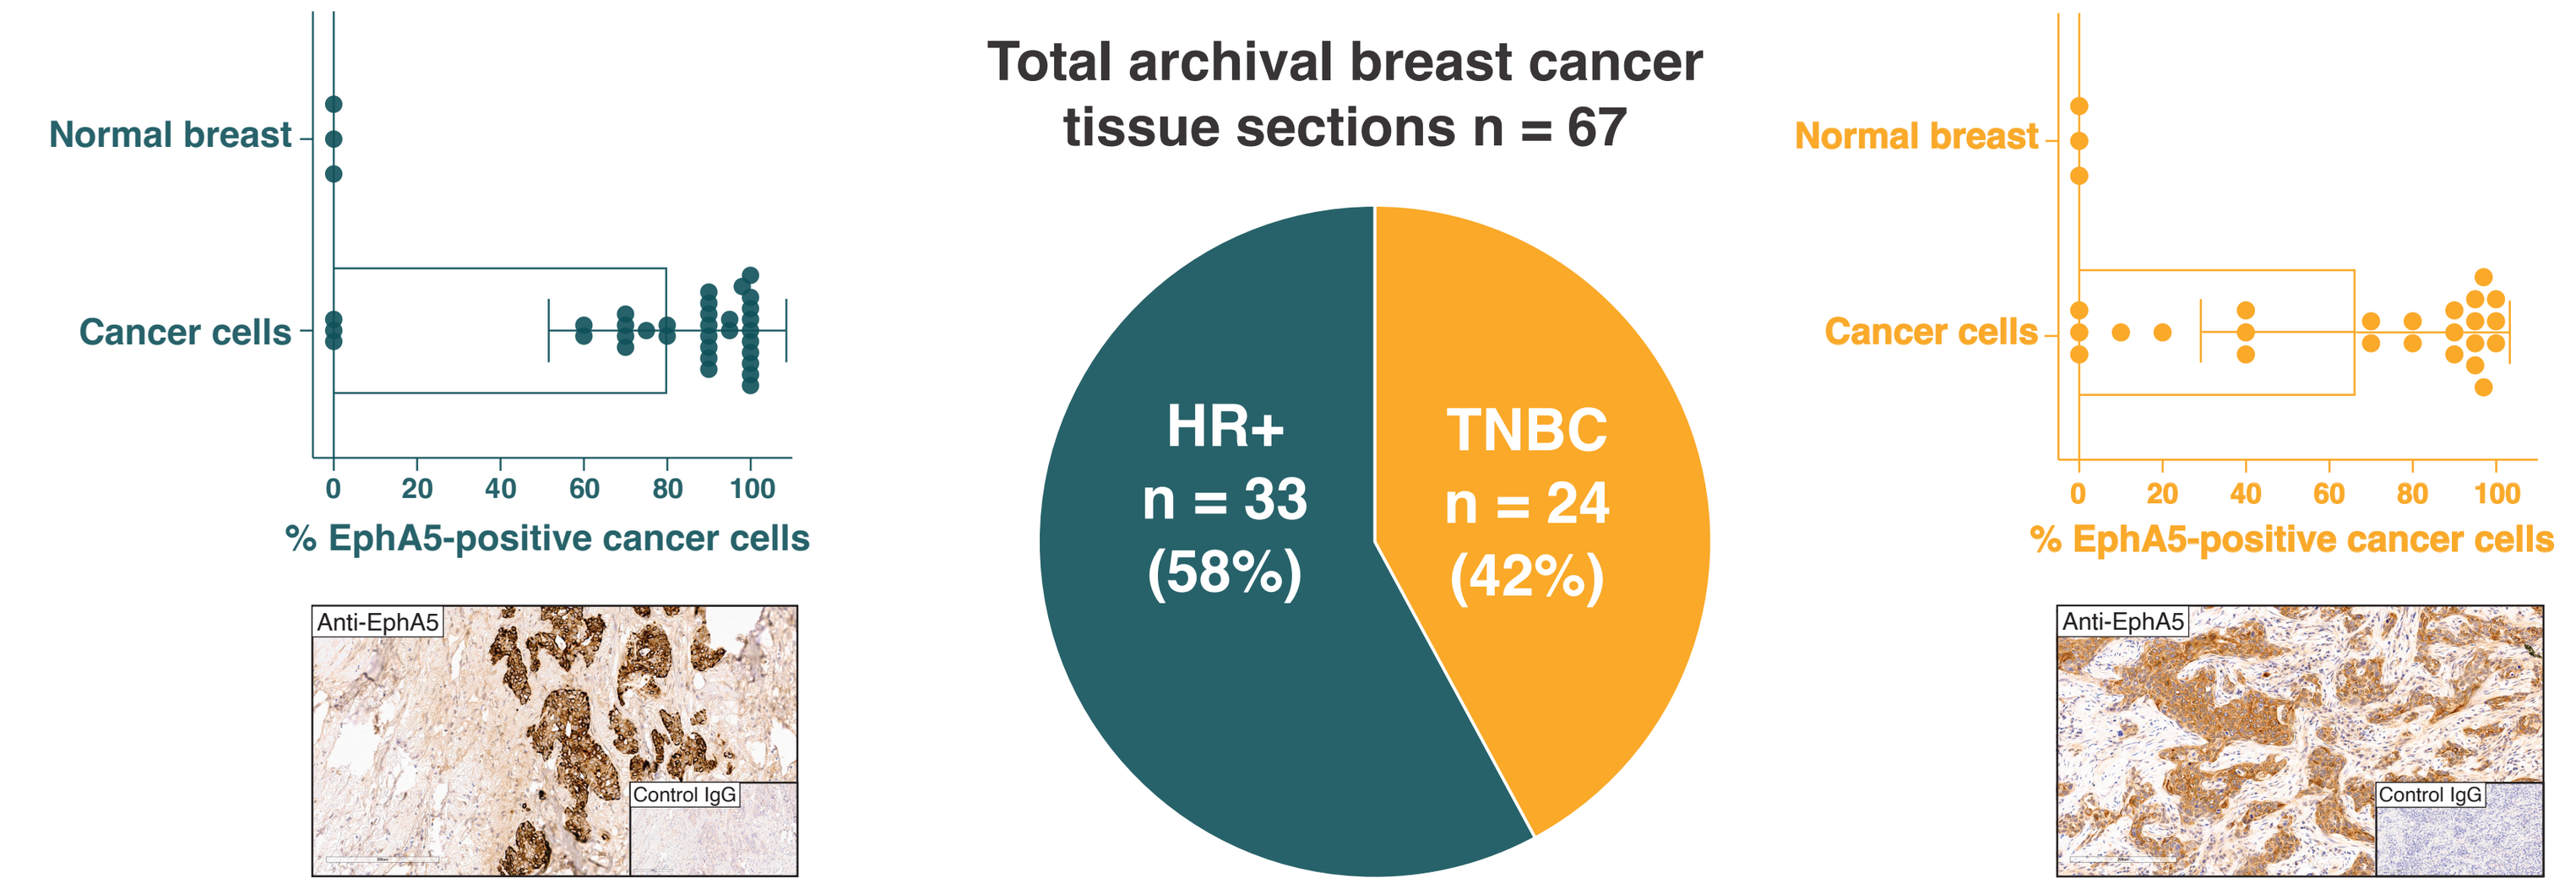
Normal breast
Cancer cells
0 20 40 60 80 100
% EphA5-positive cancer cells
Anti-EphA5
Control IgG
Total archival breast cancer tissue sections n = 67
HR+ n = 33 (58%)
TNBC n = 24 (42%)

Figure 1. Testing for EphA5 expression by IHC used a commercial rabbit polyclonal antibody. IHC staining was performed on the Leica BOND III automated platform.

MBRC-101

• MBRC-101 is composed of a humanized anti-EphA5 IgG1 antibody conjugated to MMAE

• The anti-EphA5 antibody binds to EphA5 specifically and with high affinity (KD = 2.05×10^{-9} M)

• The anti-EphA5 antibody/EphA5 complex internalizes rapidly in EphA5-expressing cells

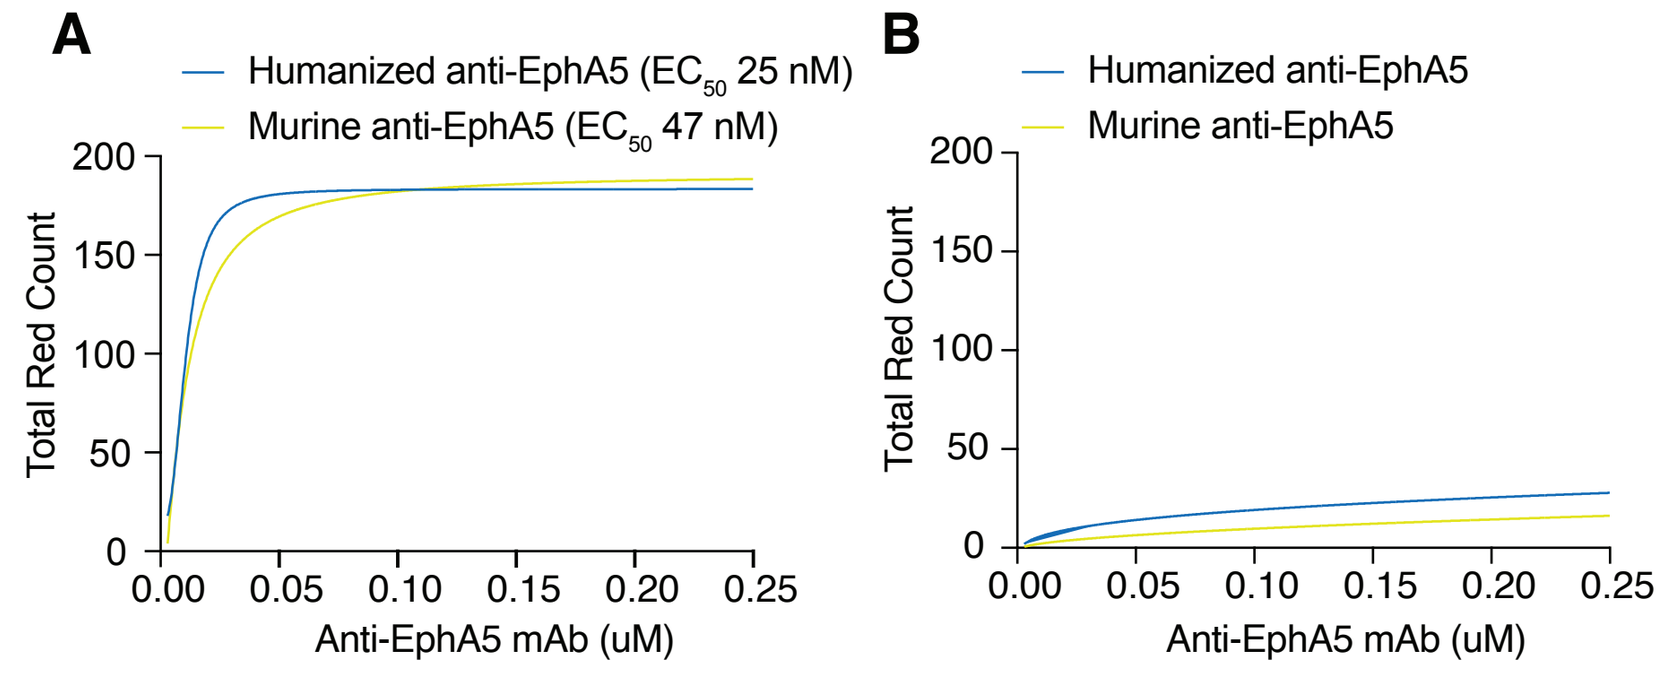
A
Humanized anti-EphA5 (EC_{50} 25 nM)
Murine anti-EphA5 (EC_{50} 47 nM)
Total Red Count
Anti-EphA5 mAb (uM)
B
Humanized anti-EphA5
Murine anti-EphA5

Figure 2. (**A**) Antibody internalization was visualized and quantified using the Incucyte® real-time live cell imaging analysis. (**B**) Antibody internalization was not detected in EphA5-negative cells.

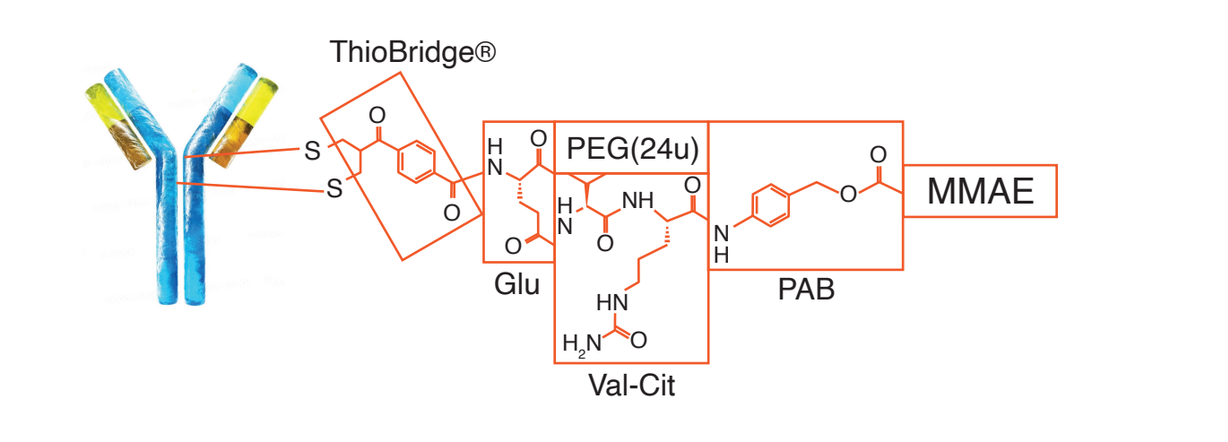
ThioBridge®
PEG(24u)
MMAE
Glu
Val-Cit
PAB

Figure 3. MBRC-101 is composed of a humanized IgG1 monoclonal antibody conjugated via a linker to MMAE. Site-directed conjugation uses the ThioBridge® technology and a protease labile linker, resulting in a linker-payload structure of ThioBridge®-Glu-[Val-Cit-PAB-MMAE] PEG[24u]).

Methods and Results

• MBRC-101 is cytotoxic to EphA5-expressing cells in a concentration-dependent manner

• MBRC-101 binding epitope is conserved in rats and non-human primates

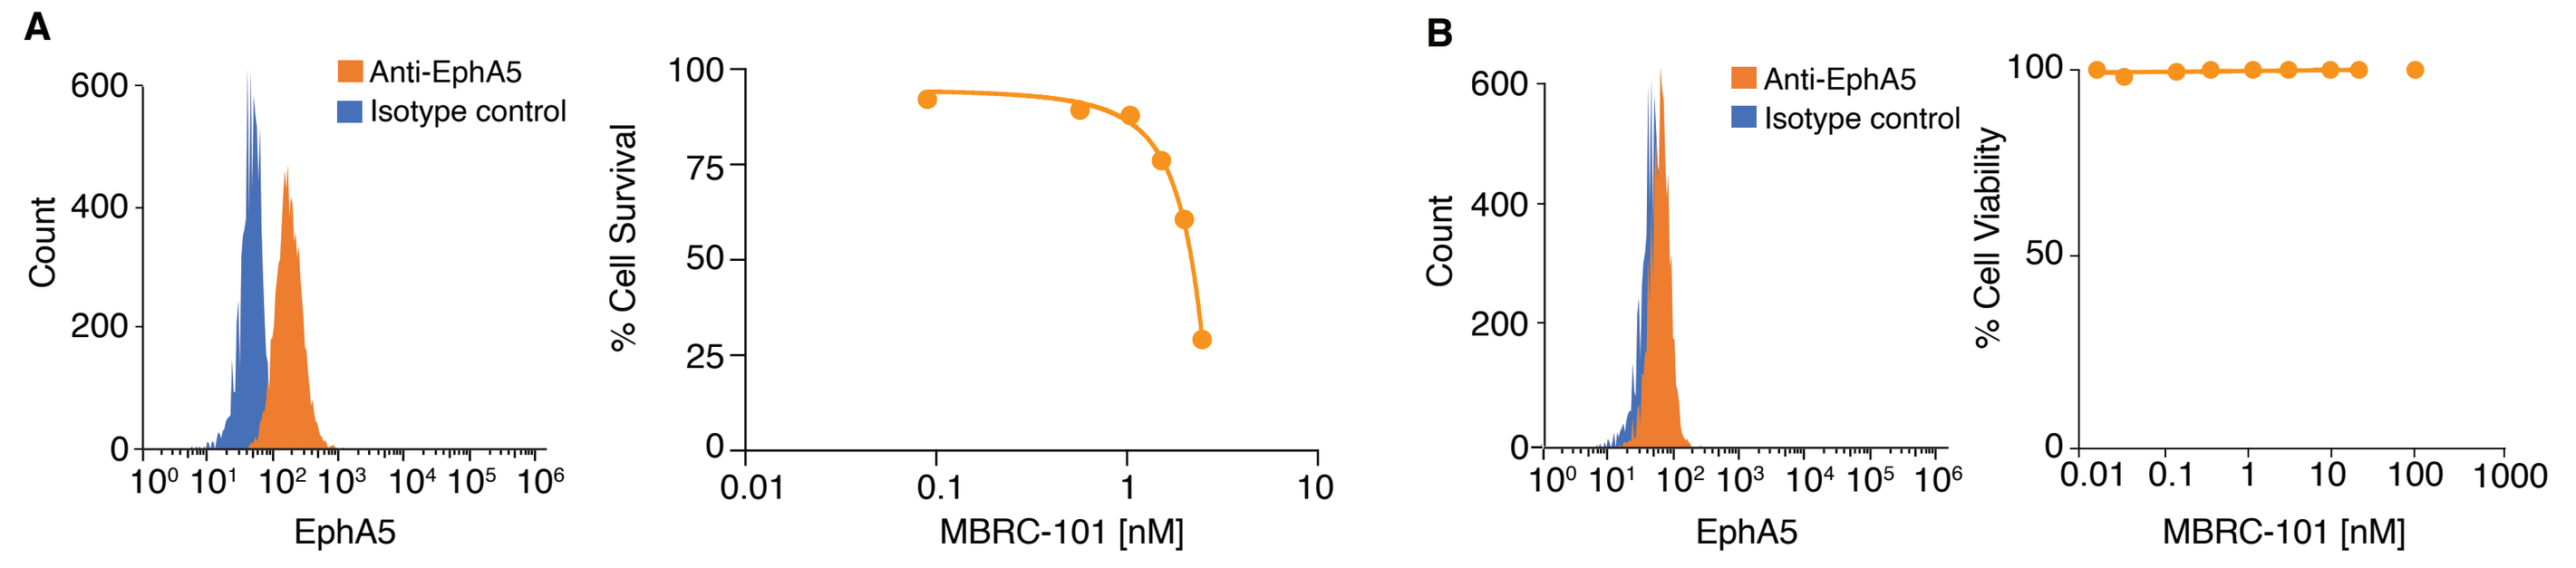
A
Anti-EphA5
Isotype control
Count
EphA5
% Cell Survival
MBRC-101 [nM]
B
% Cell Viability

Figure 4. (**A**) Flow cytometry analysis confirming EphA5 expression on cells. MBRC-101 kills EphA5-positive cells in a dose dependent manner. (**B**) Flow cytometry analysis confirming absence of EphA5 on control cells. MBRC-101 does not affect viability of control cells.

Human	$_{300}$GTCQVCRPGFFKASPHIQSCGKCPPHSYTHEEASTS$_{335}$
Rhesus monkey	$_{300}$GTCQVCRPGFFKASPHIQSCGKCPPHSYTHEEASTS$_{335}$
Cynomolgus monkey	$_{300}$GTCQVCRPGFFKASPHIQSCGKCPPHSYTHEEASTS$_{335}$
Rat	$_{300}$GTCQVCRPGFFKASPHSQTCSKCPPHSYTHEEASTS$_{335}$

Figure 5. Epitope identification by alanine scanning mutagenesis and high throughput flow cytometry. Amino acid residues essential for binding to EphA5 are highlighted in green. Amino acid residues that are not essential but contribute to binding are highlighted in yellow.

MBRC-101 is highly potent *in vivo*

• Weekly administrations of intravenous (IV) MBRC-101 showed dose-dependent, robust, and reproducible anti-tumor activity in patient-derived xenograft models of TNBC

• Partial tumor responses were observed at doses of 2.5 mg/Kg IV and complete tumor responses at 5 mg/Kg IV

• Doses up to 20 mg/Kg IV were well-tolerated with no observable weight loss

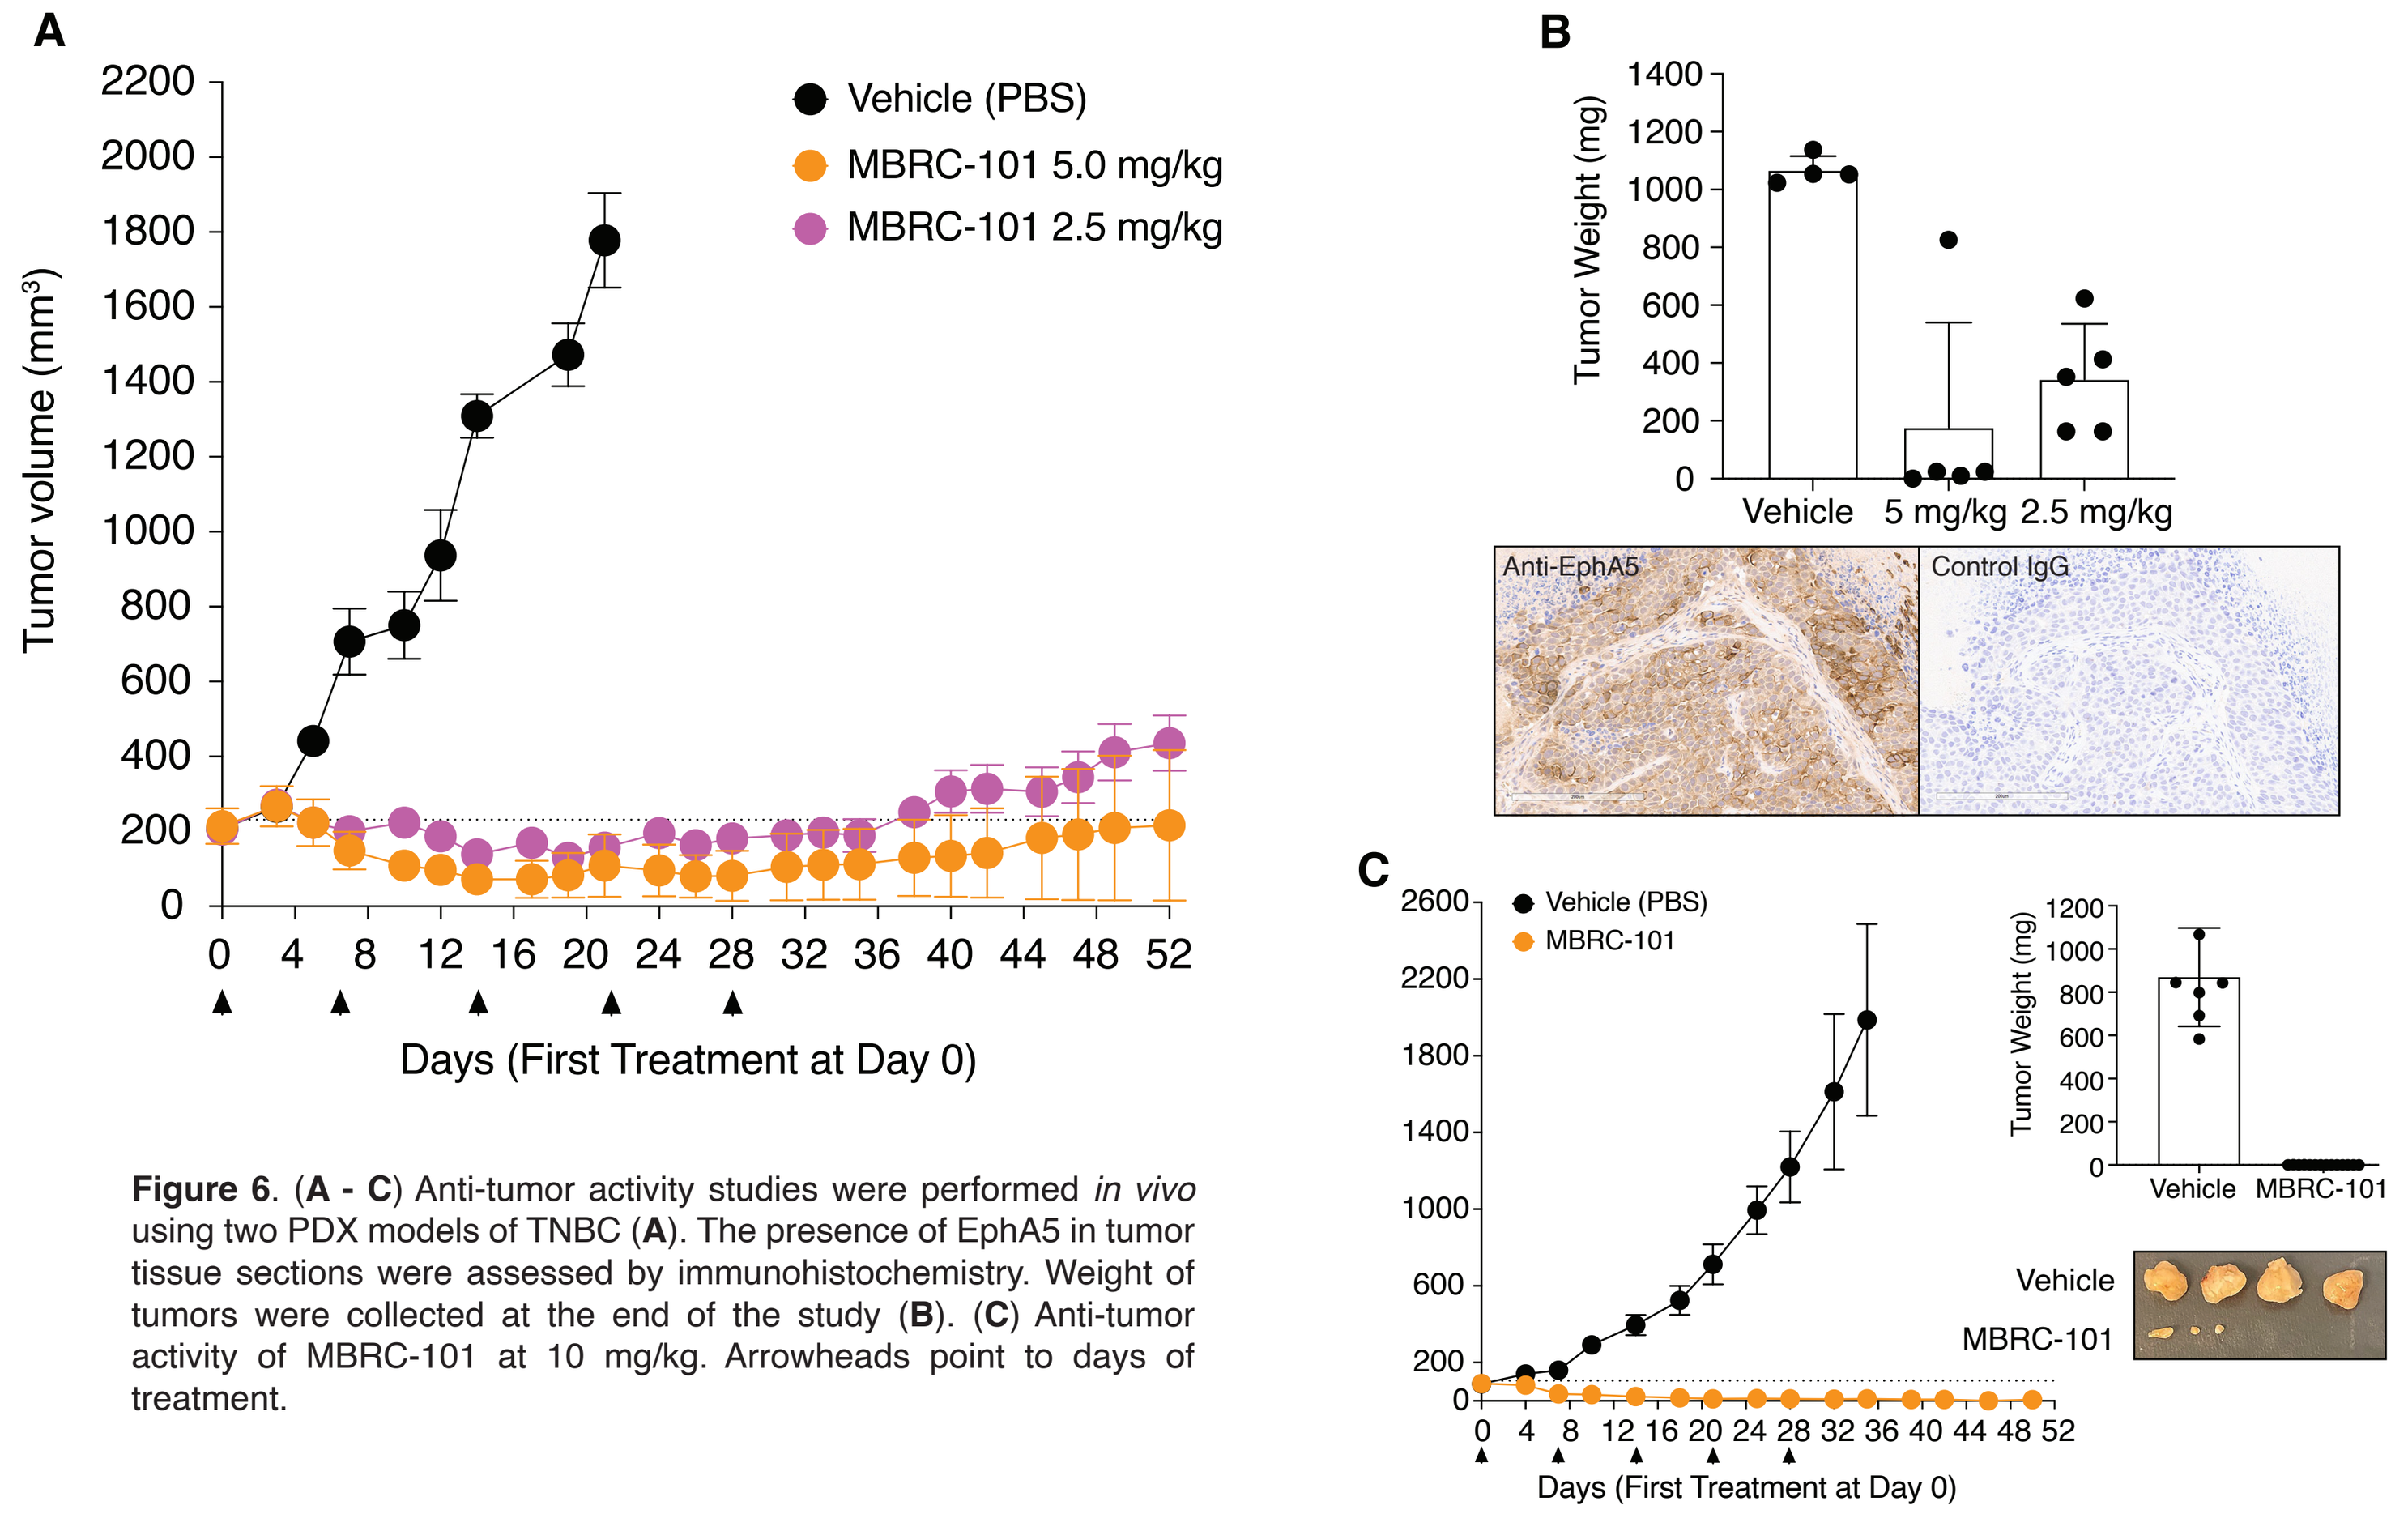
A
Vehicle (PBS)
MBRC-101 5.0 mg/kg
MBRC-101 2.5 mg/kg
Tumor volume (mm³)
Days (First Treatment at Day 0)
B
Tumor Weight (mg)
Vehicle
5 mg/kg
2.5 mg/kg
Anti-EphA5
Control IgG
C
Vehicle (PBS)
MBRC-101
Vehicle
MBRC-101

Figure 6. (**A - C**) Anti-tumor activity studies were performed *in vivo* using two PDX models of TNBC (**A**). The presence of EphA5 in tumor tissue sections were assessed by immunohistochemistry. Weight of tumors were collected at the end of the study (**B**). (**C**) Anti-tumor activity of MBRC-101 at 10 mg/kg. Arrowheads point to days of treatment.

Toxicology findings attributed to payload and not target-related

Toxicology study design for MBRC-101

Species	Cynomolgus Monkey		Sprague-Dawley Rat	
Study	DRF	GLP	DRF	GLP
Dosing	2 IV boluses	2 IV boluses	2 IV boluses	2 IV boluses
Interval	3 wk	3 wk/4 wk recovery	3 wk	3 wk/4 wk recovery
Dose (mg/kg)	0, 5, 10, 15	0, 5, 7.5, 10	0, 5, 10, 20, 30	0, 10, 20, 30
HED* (mg/kg)	1.67, 3.2, 5	1.67, 2.5, 3.2	1, 2, 3, 5	2, 3, 5
#/Group/Sex	1F/2M or 1M/2F	3 + 2 recovery	5	10 + 5 recovery

Cynomolgus monkey - toxicology findings

Dose (mg/kg)	HED* (mg/kg)	Finding	Recovery
15	5	**Morbidity:** (n=1) due to sepsis	No
10 - 15	3.2 - 5	**Testicle:** bilateral degeneration/decreased sperm **Ovary:** decreased secondary and tertiary follicles **Neutrophils:** decreased	No No Yes
5 - 15	1.67 - 5	**Lymphocytes:** decreased in blood and spleen **Red blood cell mass:** decreased	Yes Yes
10	3.2	**Cornea:** minimal bilateral degeneration at the limbus	Yes

Sprague-Dawley rat - toxicology findings

Dose (mg/kg)	HED* (mg/kg)	Finding	Recovery
20 - 30	3 - 5	**Liver** • Hepatocellular necrosis, increased ALT, AST • Altered function: increased total bilirubin, ALP, cholesterol and triglycerides (30 mg/kg only)	 Yes Yes
		Neutrophils: decreased	Yes
		Red blood cell mass: decreased	Yes
		Cornea: increased mitotic figures and apoptosis	Yes
10 - 30	2 - 5	**Lung:** increased alveolar macrophage and aveolar hyperplasia	No
		Testicle: bilateral degeneration with decreased sperm	No
		Lymphocytes: decreased in blood and thymus	Yes

*Human equivalent dose

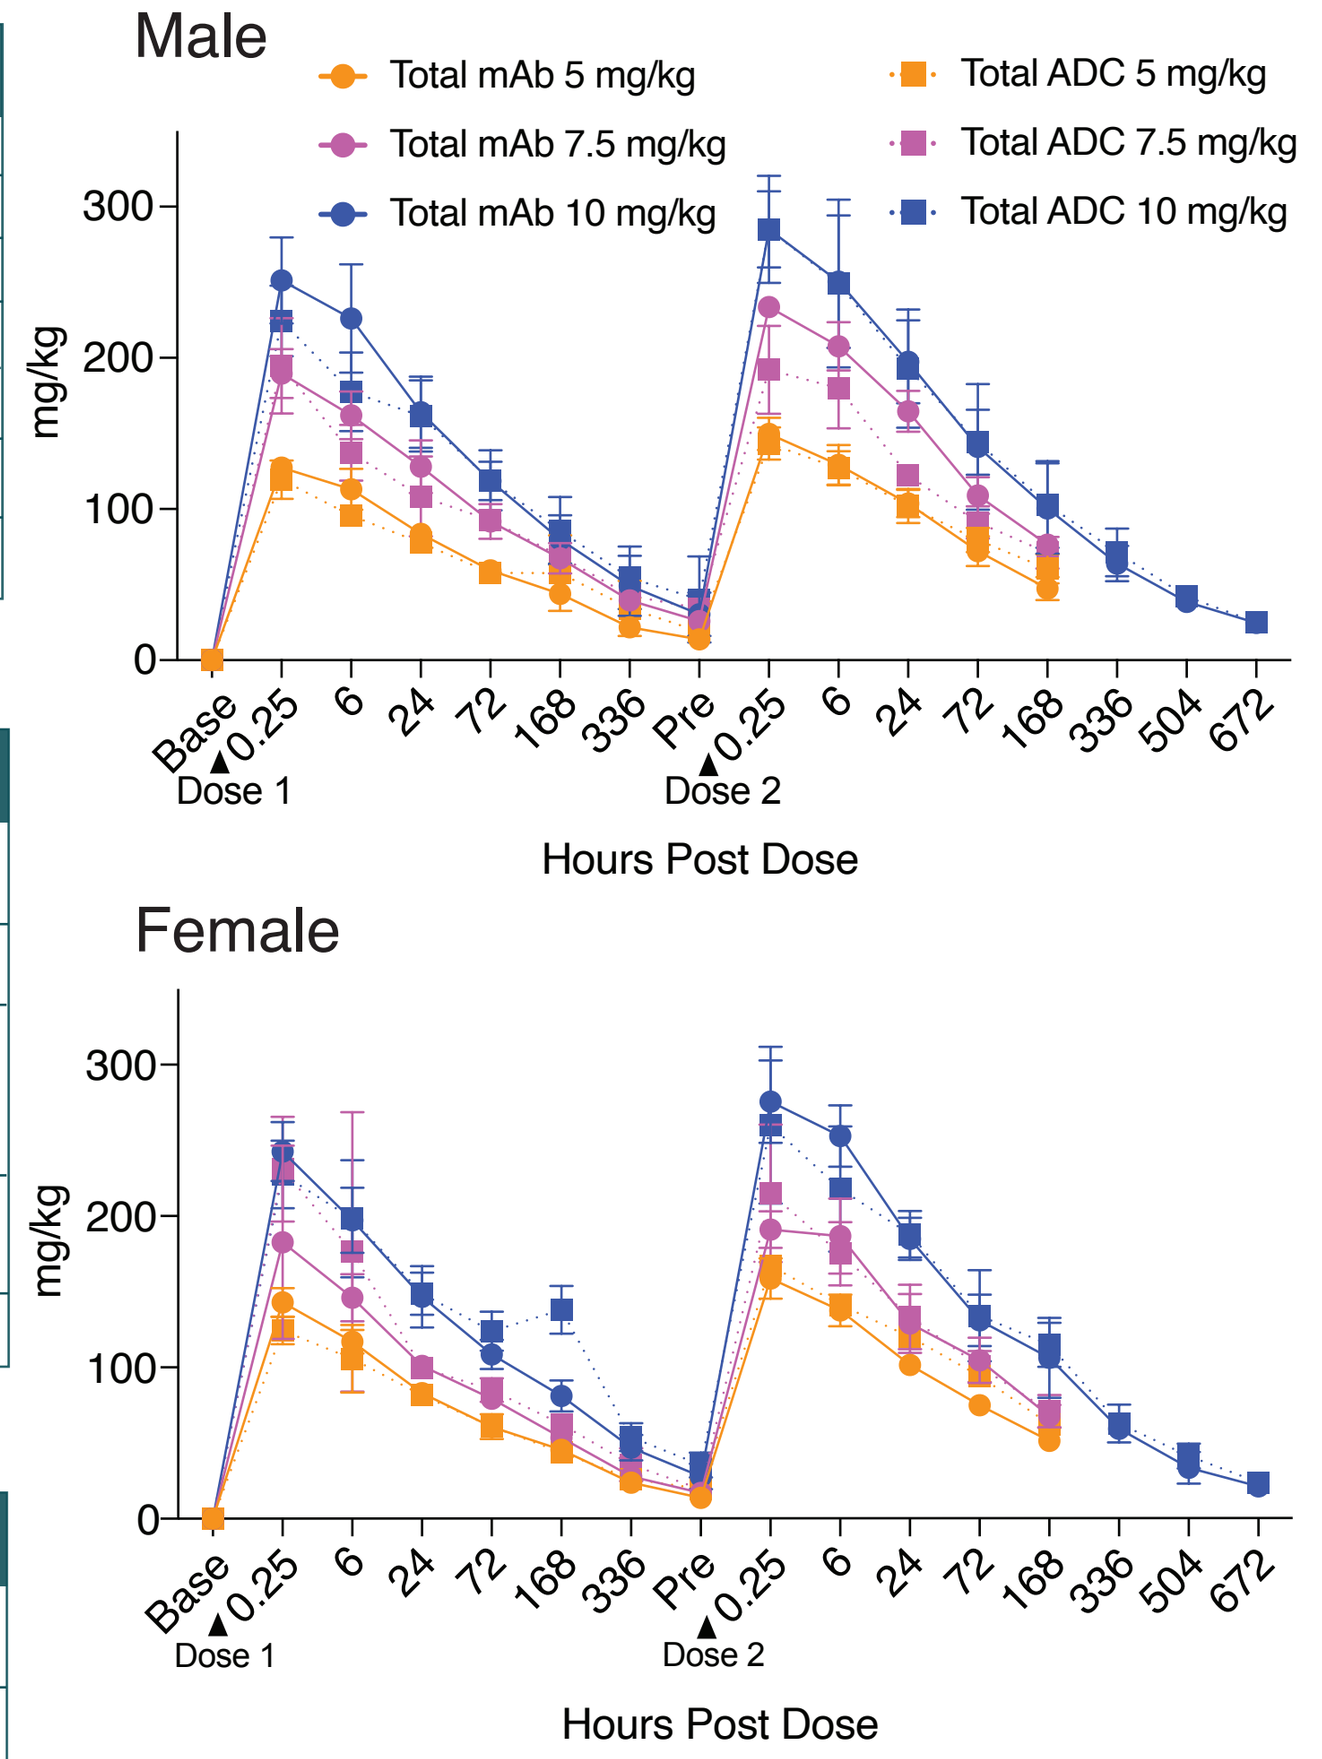
Male
Total mAb 5 mg/kg
Total mAb 7.5 mg/kg
Total mAb 10 mg/kg
Total ADC 5 mg/kg
Total ADC 7.5 mg/kg
Total ADC 10 mg/kg
mg/kg
Dose 1
Dose 2
Hours Post Dose
Female

Figure 7. Due to the stability of MBRC-101, exposure for total ADC and total antibody were highly concordant. Both sexes demonstrated similar pharmacokinetics with no accumulation in the second dose; Cmax at 0.25 hours was consistent with IV bolus.

• Half-life of MBRC-101 in cynomolgus monkeys = 7 to 11 days

• Half-life of MBRC-101 in rats = 8 to 20 days

• Highly stable linker leads to equivalent levels of total ADC and total antibody with very low levels of free MMAE (< 0.01 %)

MBRC-101-001 Phase 1/1b: Study Schema

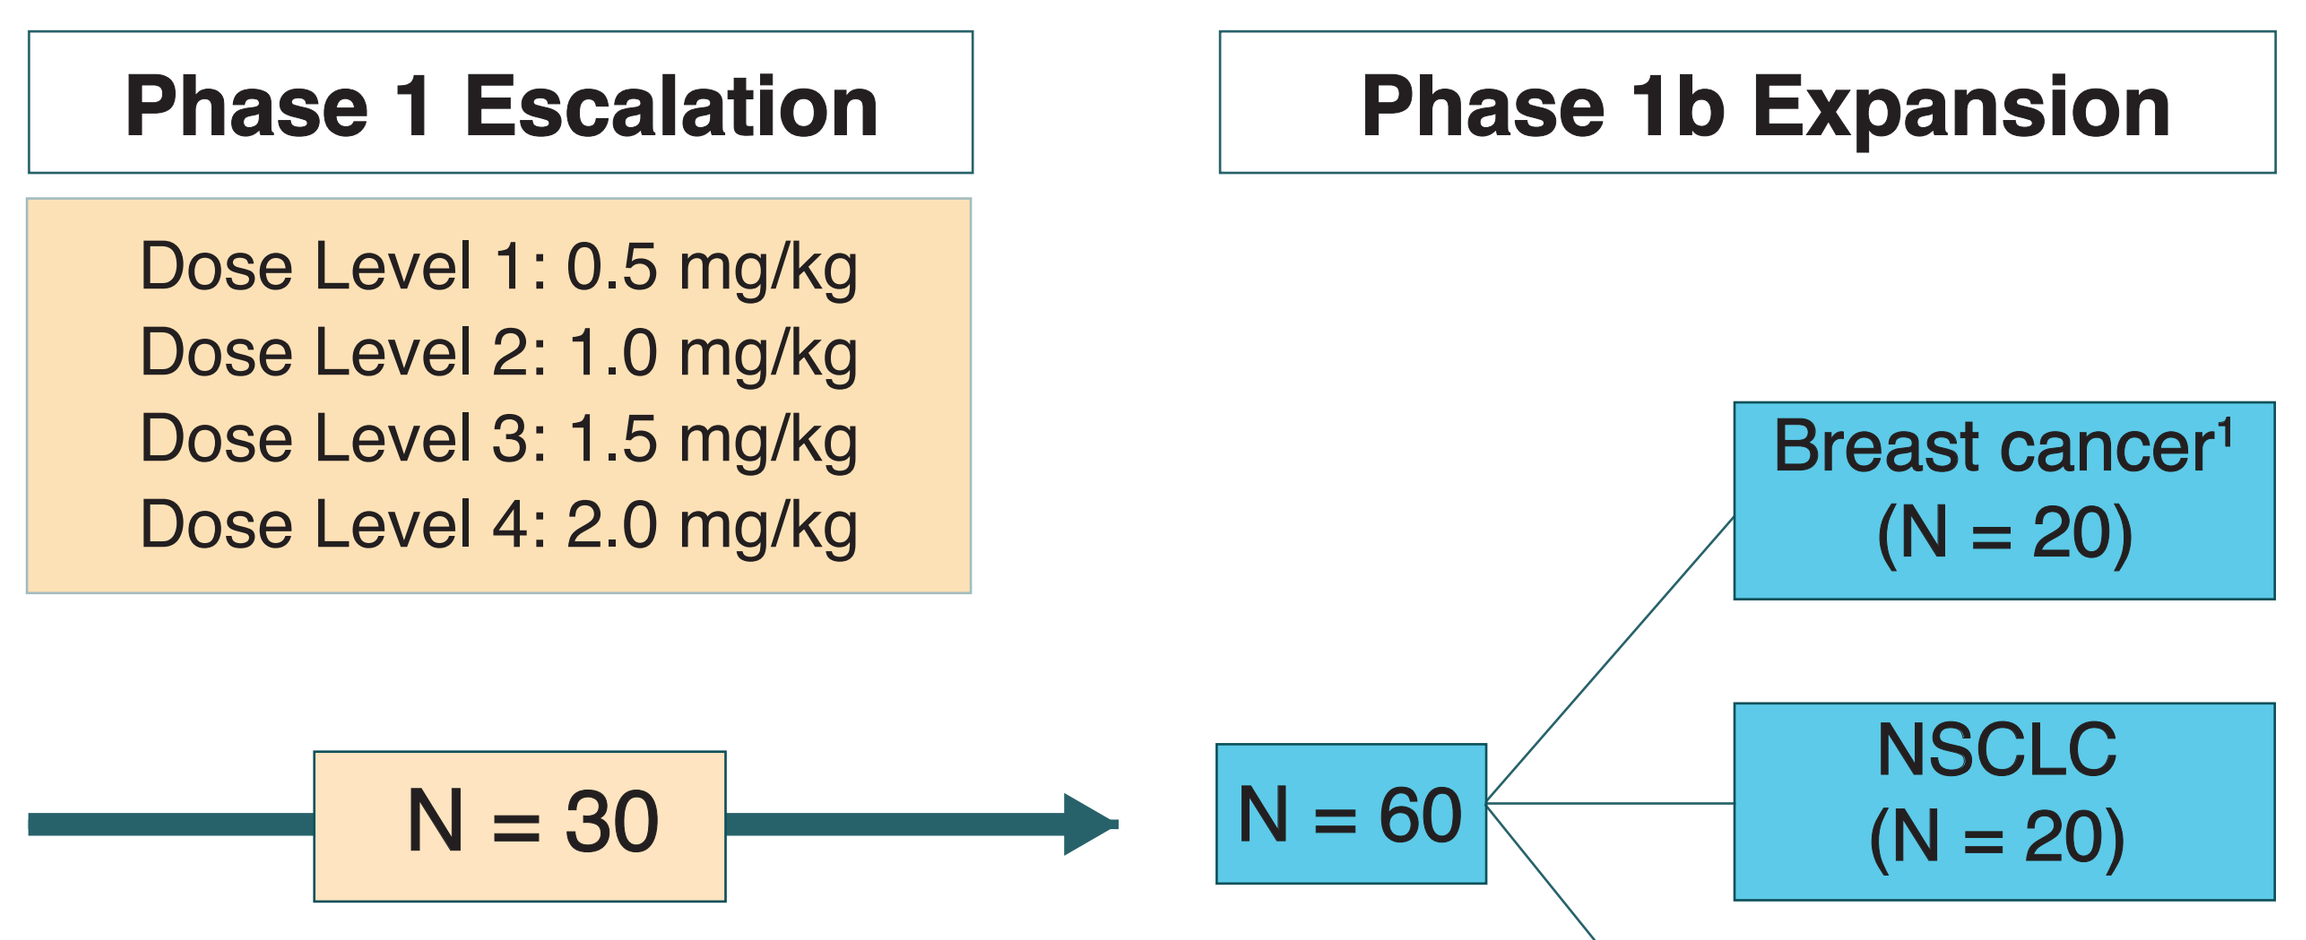
Phase 1 Escalation
Dose Level 1: 0.5 mg/kg
Dose Level 2: 1.0 mg/kg
Dose Level 3: 1.5 mg/kg
Dose Level 4: 2.0 mg/kg
N = 30
Phase 1b Expansion
N = 60
Breast cancer¹ (N = 20)
NSCLC (N = 20)
Tumor agnostic² (N = 20)

NSCLC: Non-small cell lung cancer

[1]Triple negative or HR+/HR-

[2]EphA5+ by IHC analysis

Phase 1/1b Trial (Cont.)

Key Eligibility Criteria

• Histologic or cytologic diagnosis of malignant solid tumor

• Refractory to conventional therapy

• No requirements for prior lines of therapy

• Prior ADC therapy is allowed

• Prior MMAE-based ADCs are allowed for Phase 1 but not Phase 1b

• EphA5 expression by IHC required only for Phase 1b tumor agnostic cohort

Ab = total antibody; **AE** = adverse event; **DLT** = dose limiting toxicity; **DOR** = duration of response; **IHC** = immunohistochemistry; **MMAE** = monomethyl auristatin E; **MTD** = maximum tolerated dose; **OBRD** = optimal biologically relevant dose; **ORR** = overall response rate; **PFS** = progression free survival; **PK** = pharmacokinetics; **RECIST** = Response Evaluation Criteria in Solid Tumors; **RP2D** = recommended Phase 2 doses; **SAE** = serious adverse event

Objectives	End points
Phase 1 Primary Objectives and Endpoints	
• To identify potential OBRD and dosing regimens • To establish the MTD of MBRC-101 using one or more dose regimens • To identify potential RP2Ds and regimens of MBRC-101	• MTD, DLTs, AEs, SAEs, and clinical laboratory tests
Phase 1b	
• To evaluate the safety of MBRC-101 at potential OBRDs, RP2Ds and dosing regimens	• AEs, SAEs, and clinical laboratory tests
• To evaluate preliminary clinical activity of MBRC-101 at potential OBRDs and dosing regimens	• Investigator-assessed ORR by response RECIST v1.1
Phase 1 and 1b Secondary Objectives and Endpoints	
• To characterize single and multiple dose PK profiles	• MBRC-101, Ab, and unconjugated MMAE blood concentrations
• To evaluate incidence and persistence of anti-MBRC-101 Ab formation	• Anti-MBRC-101 Ab blood concentration
• To evaluate biomarkers of clinical response and resistance, safety, pharmacodynamic activity, and/or mechanism of action	• EphA5 expression as determined by IHC

Conclusions

• Extensive *in vitro* testing showed that MBRC-101 binds to EphA5 exclusively, it is rapidly internalized and is cytotoxic to cells expressing EphA5

• MBRC-101 is highly potent against PDX models of TNBC

• MBRC-101 was well tolerated at doses up to 30 mg/kg in rats and 10 mg/kg in monkeys. Toxicologic findings in both species were attributed to the MMAE payload and not target-related

• Highest non-severely toxic dose (HNSTD) in monkeys was 10 mg/kg (HED = 3.2 mg/kg). In alignment with ICH S9, 1/6 of the monkey HNSTD was used to determine a human start dose of 0.5 mg/kg

• At 0.5 mg/kg the safety margin was approximated at 15-fold based on projected human exposures

• A Phase 1/1b trial of MBRC-101 is open and currently enrolling patients (NCT06014658)

References

1. Taylor V., et al. (1994) Expression and developmental regulation of Ehk-1, a neuronal Elk-like receptor tyrosine kinase in brain. Neuroscience 63, 163–178

2. Zhou R. (1997) Regulation of topographic projection by the Eph family receptor Bsk (EphA5) and its ligands. Cell Tissue Res. 290, 251–259

3. Murai K. K. & Pasquale E. B. (2002) Can Eph receptors stimulate the mind? Neuron 33, 159–162

4. Giaginis C., et al. (2010) Clinical significance of ephrin (eph)-A1, -A2, -a4, -a5 and -a7 receptors in pancreatic ductal adenocarcinoma. Pathol. Oncol. Res. 16, 267–276

5. Pejovic T., et al. (2009) Expression profiling of the ovarian surface kinome reveals candidate genes for early neoplastic changes. Transl. Oncol. 2, 341–349

6. Zhang, W., et al. (2019). Differential expression of EphA5 protein in gastric carcinoma and its clinical significance. Oncology letters, 17(6), 5147–5153

7. Staquicini, F. I., et al. (2015). Receptor tyrosine kinase EphA5 is a functional molecular target in human lung cancer. The Journal of Biological Chemistry, 290(12), 7345–7359

8. D'Angelo, S., Staquicini, F. I., et al. (2018). Selection of phage-displayed accessible recombinant targeted antibodies (SPARTA): methodology and applications. JCI insight, 3(9), e98305

Contact

Fernanda I. Staquicini, Ph.D.
Director of Research and Development
MBrace Therapeutics, Inc.
fstaquicini@mbracetrx.com

J. Kellogg Parsons, M.D.
Vice-President of Clinical Development
MBrace Therapeutics, Inc.
kparsons@mbracetrx.com

This presentation is the intellectual property of the author/presenter. Contact them at fstaquicini@mbracetrx.com for permission to reprint and/or distribute
Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster